PSMA x CD3 T-cell engagers show preclinical efficacy for the treatment of prostate cancer

AUTHORS
Peter Bergqvist,* Alaa Amash, Girija Bodhankar, Kelly Bullock, Lauren Clifford, Patrick Farber, Jessica Fernandes Scortecci, Karine Herve, Michael Kennedy, Ingrid Knarston, Tallie Kuang, Ahn Lee, Cindy-Lee Crichlow, Franco Li, Matt Mai, Stephanie K. Masterman, Melanie Pieber, Janice Reimer, Eduardo Solano Salgado, Raffi Tonikian, Gesa Volkers, Christopher Williamson, Allison Goodman, Amy C.H. Lee, Lindsay DeVorkin.

*presenter

AUTHOR AFFILIATION
AbCellera, Vancouver, Canada

DOWNLOAD POSTER

6012

BACKGROUND

PSMA-targeted T-cell engagers for the treatment of prostate cancer

Metastatic castration-resistant prostate cancer (mCRPC) is an aggressive disease that presents high unmet need, with current therapies unable to generate durable responses in most patients and a five-year relative survival rate of ~30%.[1]

Prostate-specific membrane antigen (PSMA) is a validated target for mCRPC that is being prosecuted using several modalities in the clinic.[2] PSMA x CD3 T-cell engagers (TCEs) have shown promise in preclinical and early clinical studies,[3] yet generating molecules with a therapeutic window to support efficacious dosing in patients has been a barrier to development.

AIM

Generate PSMA-targeted TCEs with sustained tumor-cell killing and *in vivo* anti-tumor efficacy

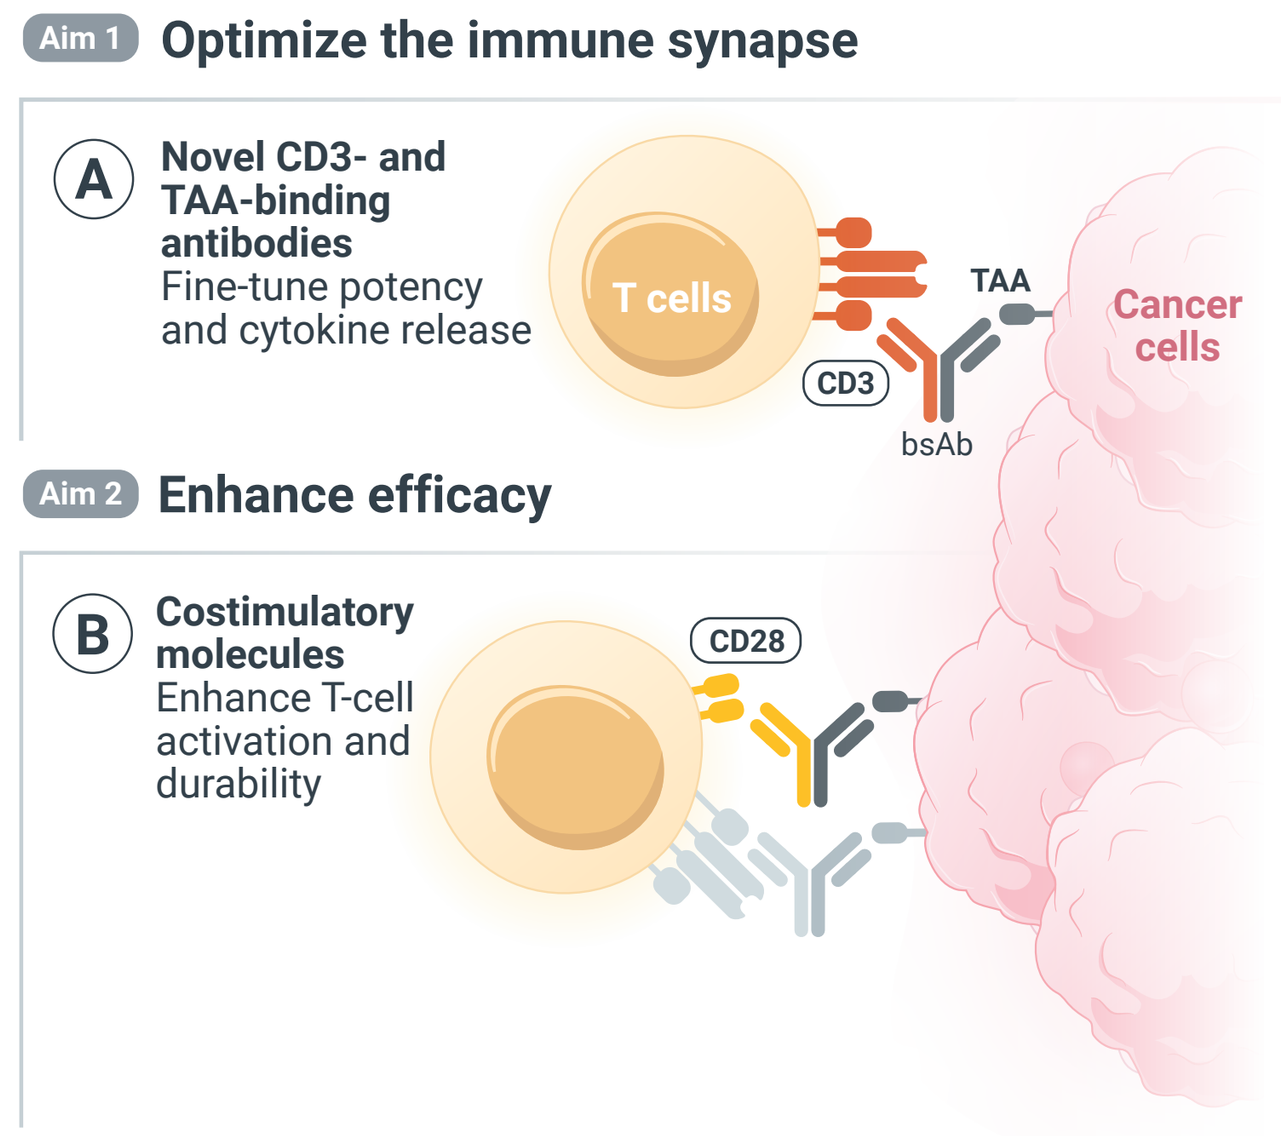
Aim 1 Optimize the immune synapse
A Novel CD3- and TAA-binding antibodies
Fine-tune potency and cytokine release
T cells
CD3
bsAb
TAA
Cancer cells
Aim 2 Enhance efficacy
B Costimulatory molecules
Enhance T-cell activation and durability
CD28

Figure 1. Strategies for building optimized TCEs. **(A)** Optimal pairing of diverse CD3- and tumor-associated antigen (TAA)-binding antibodies to achieve high potency. **(B)** Costimulatory CD28-binding antibodies to further enhance anti-tumor activity of T-cell activating therapies as needed depending on target and tumor type.

Our aim was to develop PSMA x CD3 TCEs that achieve sustained efficacy with a manageable safety profile.

Our approach is to optimize components of the immune synapse to achieve a robust interaction between T cell and tumor cell (Figure 1). We alter TCE parameters that impact function, including binding affinities, geometries, and epitopes, for both CD3 and TAAs. We then apply a suite of high-throughout assessments to empirically test molecules for desired properties. To further enhance efficacy in select targets and indications, we assess TCE function with the addition of costimulatory molecules.

REFERENCES
1. National Cancer Institute. (2022). Surveillance, Epidemiology, and End Results Program. Cancer Stat Facts: Prostate Cancer, Bethesda, MD, USA. https://seer.cancer.gov/statfacts/html/prost.html
2. Giraudet A, et al. (2021). *Ther Adv Med Oncol.* 13. doi: 10.1177/17588359211053898.
3. Simão DC, et al. (2023). *Cancers (Basel).* 15(5):1412. doi: 10.3390/cancers15051412.

COPYRIGHT © ABCELLERA

TCE function is determined by both CD3- and PSMA-binding properties

A large panel of bispecific antibodies was assessed to identify the optimal epitope and affinity for TCE function

(A) PSMA x CD3 discovery and downselection

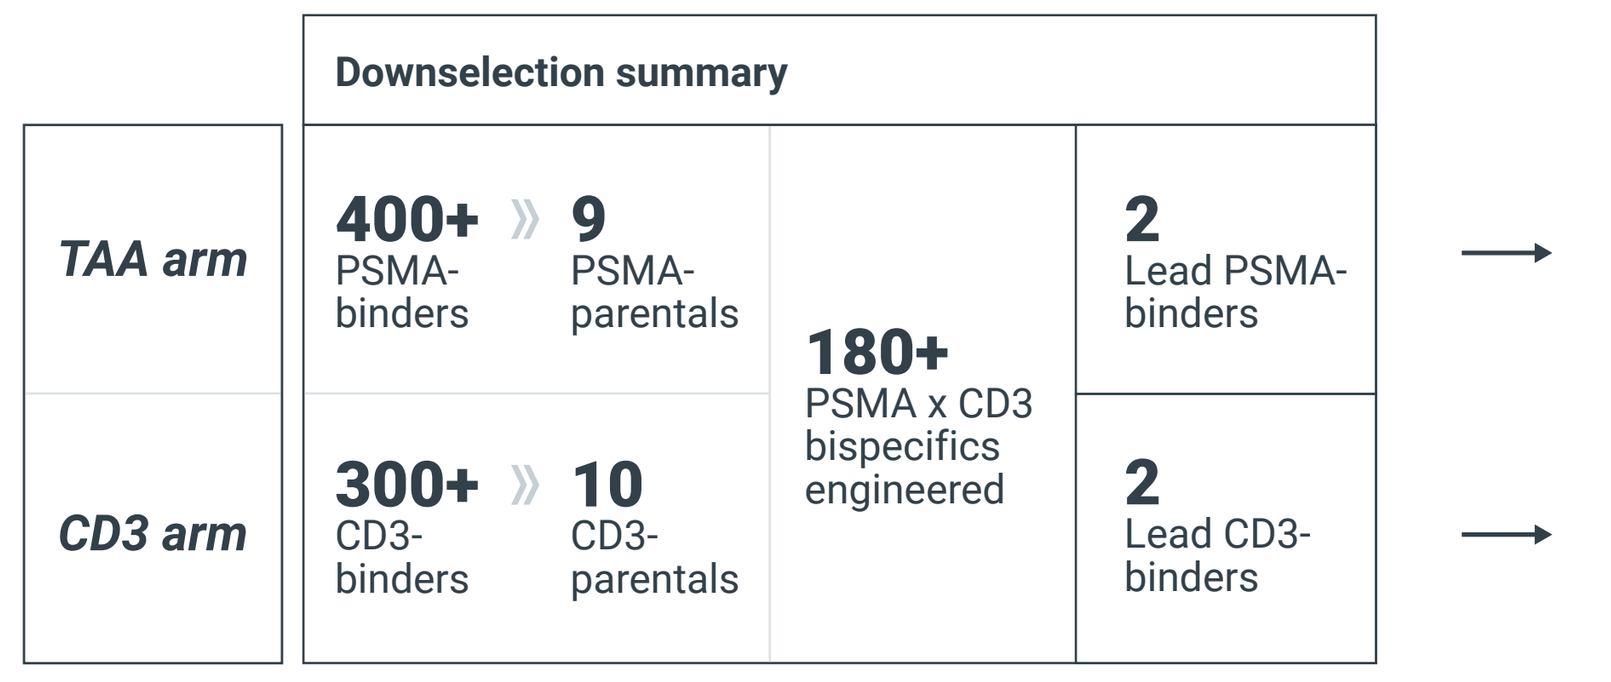
Downselection summary
TAA arm
CD3 arm
400+ PSMA-binders
9 PSMA-parentals
300+ CD3-binders
10 CD3-parentals
180+ PSMA x CD3 bispecifics engineered
2 Lead PSMA-binders
2 Lead CD3-binders

(B) Selected PSMA x CD3 bispecifics

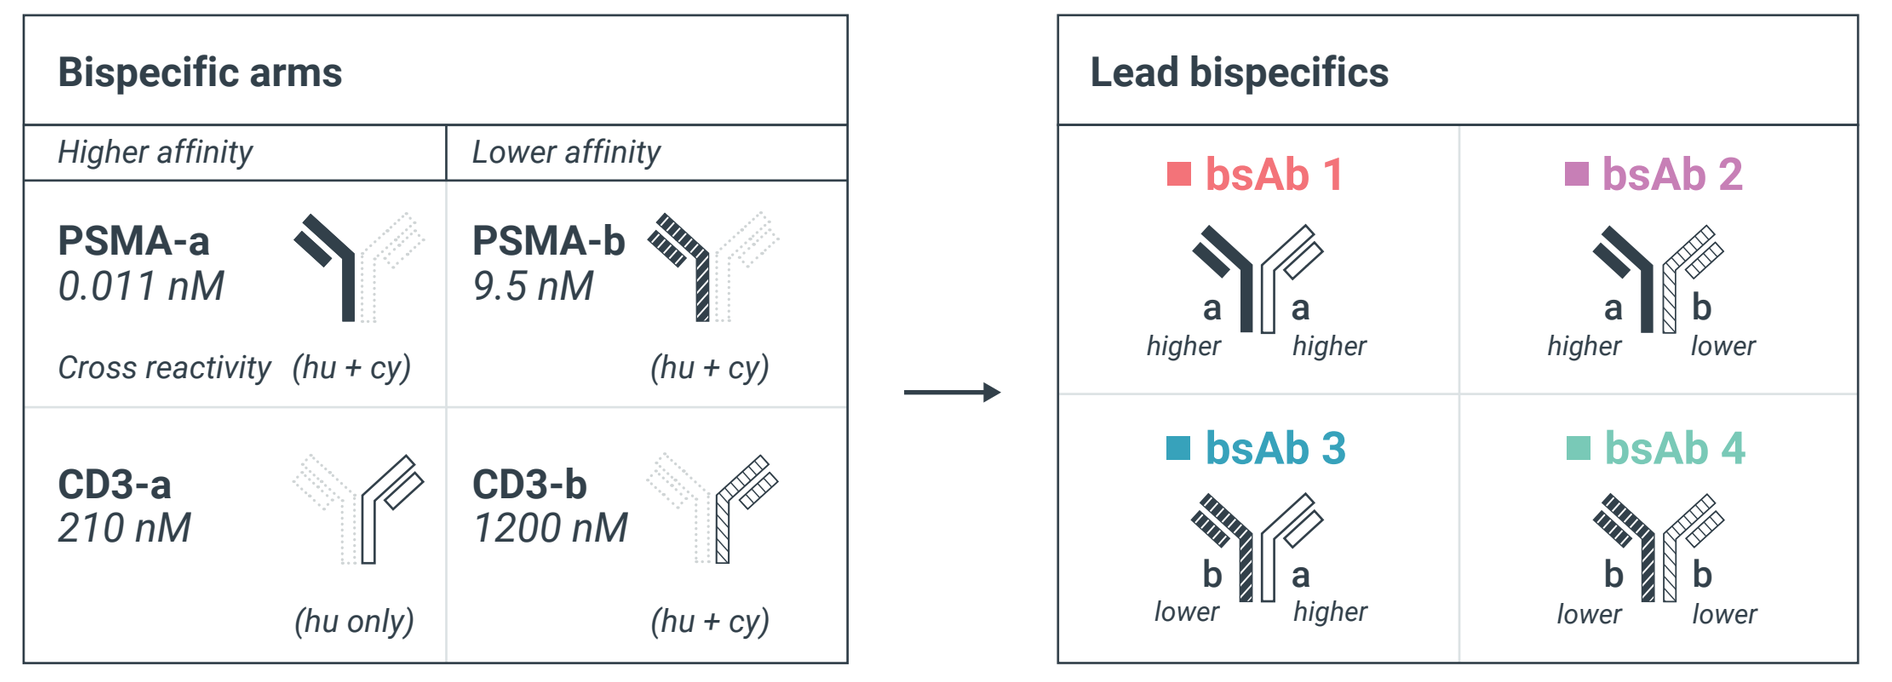
Bispecific arms
Higher affinity
Lower affinity
PSMA-a 0.011 nM
PSMA-b 9.5 nM
Cross reactivity (hu + cy)
(hu + cy)
CD3-a 210 nM
CD3-b 1200 nM
(hu only)
(hu + cy)
Lead bispecifics
bsAb 1
bsAb 2
bsAb 3
bsAb 4
higher
lower

(C) Selected CD3s bind epitopes distinct from SP34-2

High-throughput antibody discovery

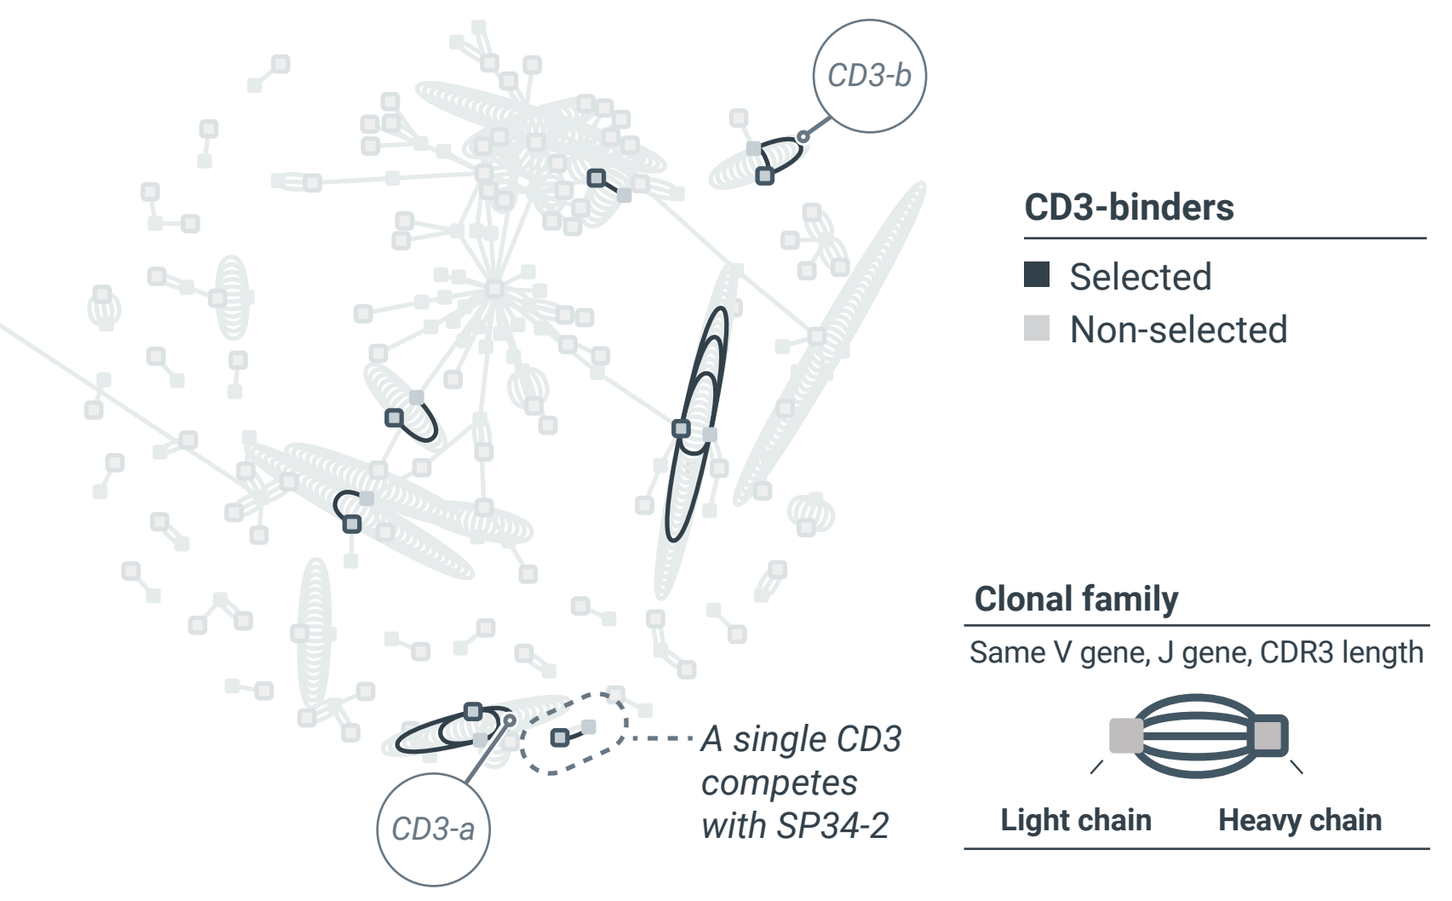
CD3-b
CD3-a
A single CD3 competes with SP34-2
CD3-binders
Selected
Non-selected
Clonal family
Same V gene, J gene, CDR3 length
Light chain
Heavy chain

(D) TAA arm bound to PSMA

Cryo-EM structure of Fab domains of PSMA x CD3 bsAbs

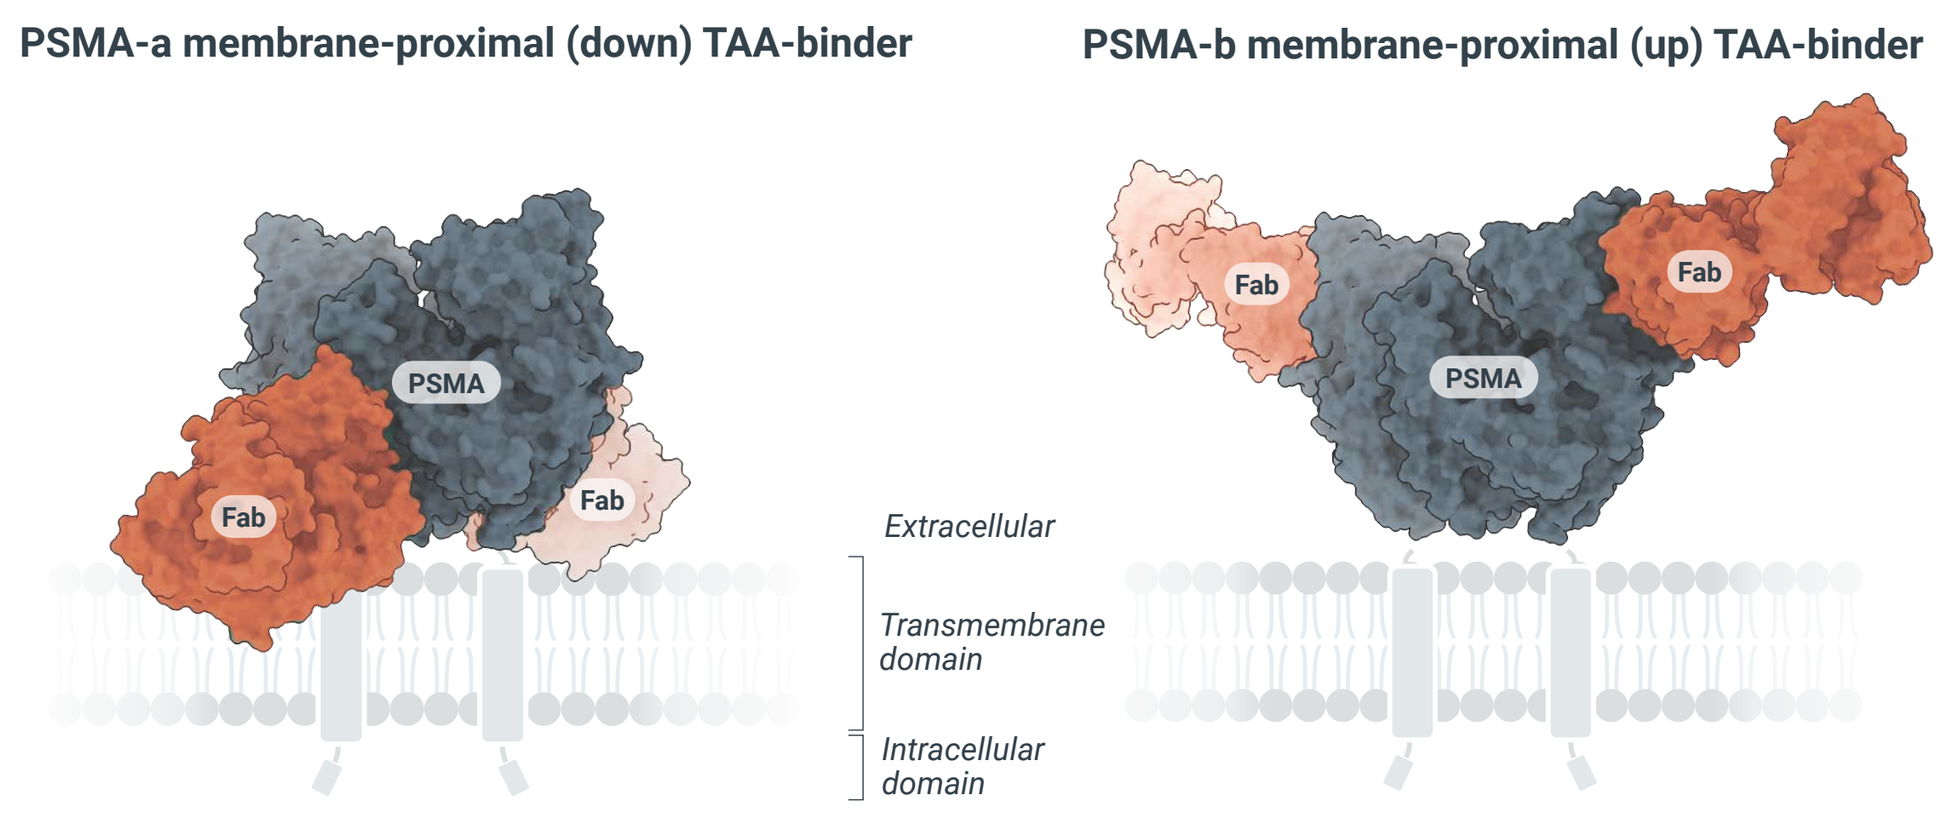
PSMA-a membrane-proximal (down) TAA-binder
PSMA-b membrane-proximal (up) TAA-binder
PSMA
Fab
Extracellular
Transmembrane domain
Intracellular domain

Figure 2. A 2x2 matrix of PSMA x CD3 bispecifics (bsAbs) to investigate structure-activity relationships. (A) Lead CD3- and PSMA-binders were selected following engineering and screening of diverse PSMA x CD3 bsAbs. **(B)** bsAbs 1-4 are comprised of two CD3- and two PSMA-binding arms to investigate the effects of epitope and affinity on TCE function. **(C)** The CD3 arms of bsAbs 1-4 bind epitopes that are distinct from that of SP34-2. AbCellera's diverse CD3 panel is visualized using Celium™. **(D)** PSMA-binding arms show membrane-proximal binding in distinct orientations, targeting distinct epitopes. Antibody-antigen complex structures were generated using a size-exclusion chromatography-purified complex and cryo-electron microscopy.

PSMA x CD3 bispecifics show sustained tumor-cell killing *in vitro*

TCEs with unique CD3- and PSMA-binding arms show varying functional profiles

(A) Tumor cell-killing and cytokine release

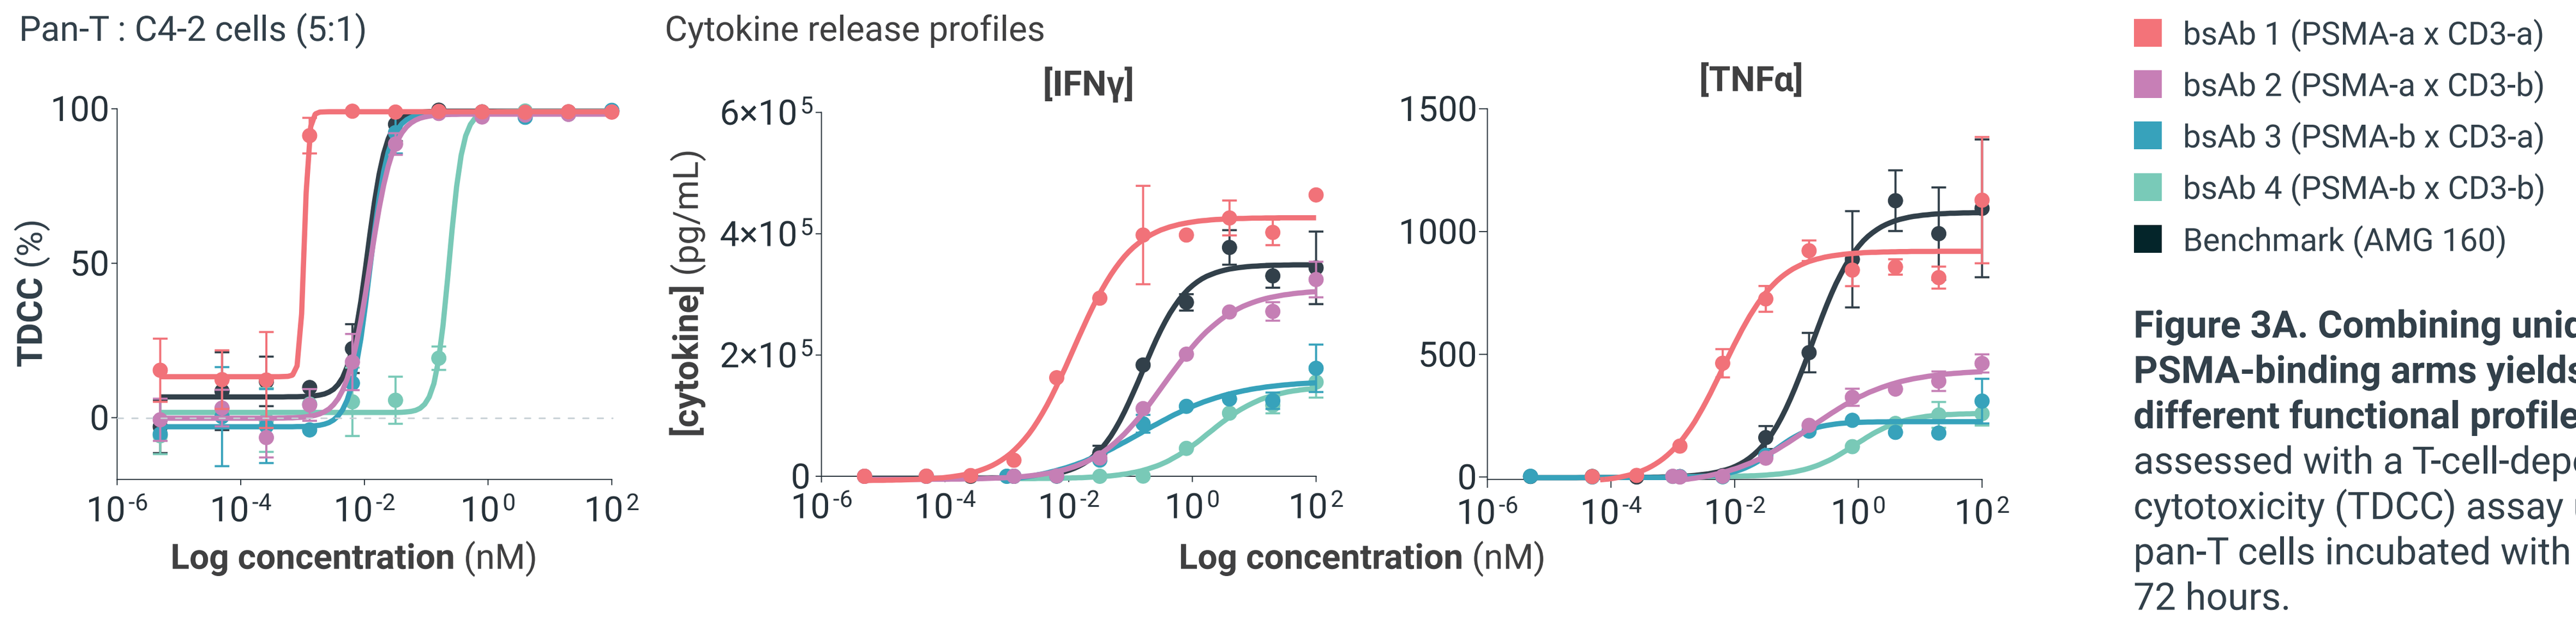
Pan-T : C4-2 cells (5:1)
TDCC (%)
Log concentration (nM)
Cytokine release profiles
[IFNγ]
[TNFα]
[cytokine] (pg/mL)
Bispecifics
bsAb 1 (PSMA-a x CD3-a)
bsAb 2 (PSMA-a x CD3-b)
bsAb 3 (PSMA-b x CD3-a)
bsAb 4 (PSMA-b x CD3-b)
Benchmark (AMG 160)

Figure 3A. Combining unique CD3- and PSMA-binding arms yields molecules with different functional profiles. Function was assessed with a T-cell-dependent cellular cytotoxicity (TDCC) assay using human pan-T cells incubated with C4-2 cells for 72 hours.

(B) Serial T-cell killing

In vitro experiment schematic

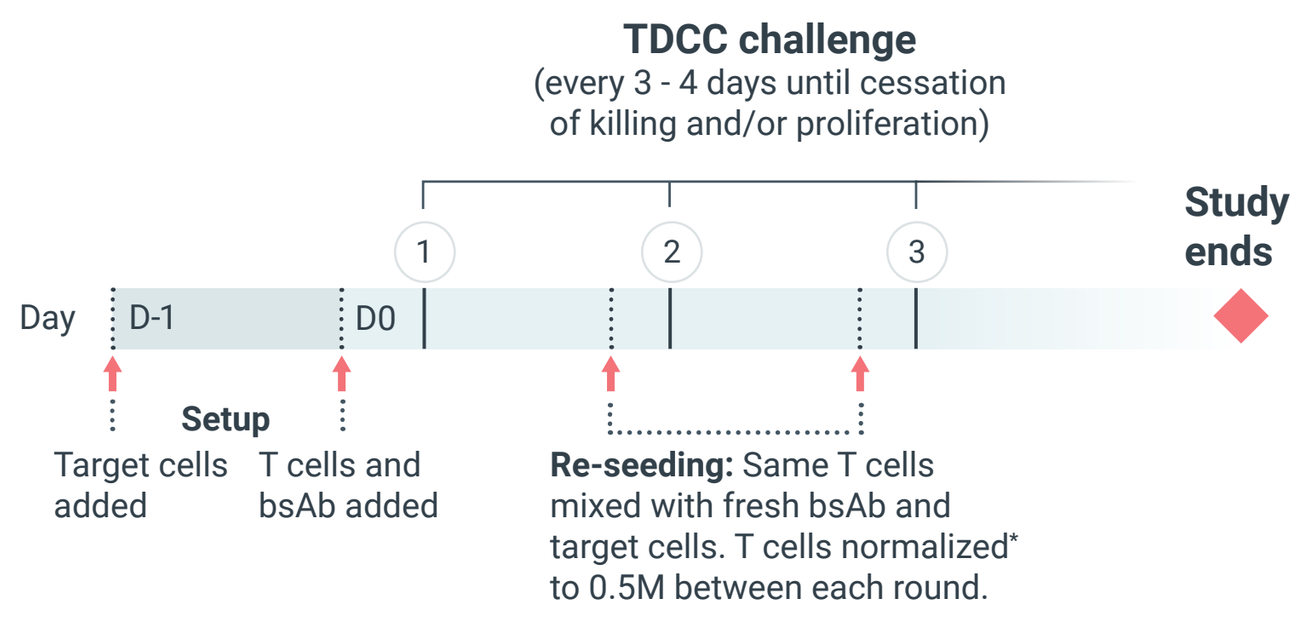
TDCC challenge
(every 3 - 4 days until cessation of killing and/or proliferation)
Study ends
Day
D-1
D0
Setup
Target cells added
T cells and bsAb added
Re-seeding: Same T cells mixed with fresh bsAb and target cells. T cells normalized* to 0.5M between each round.

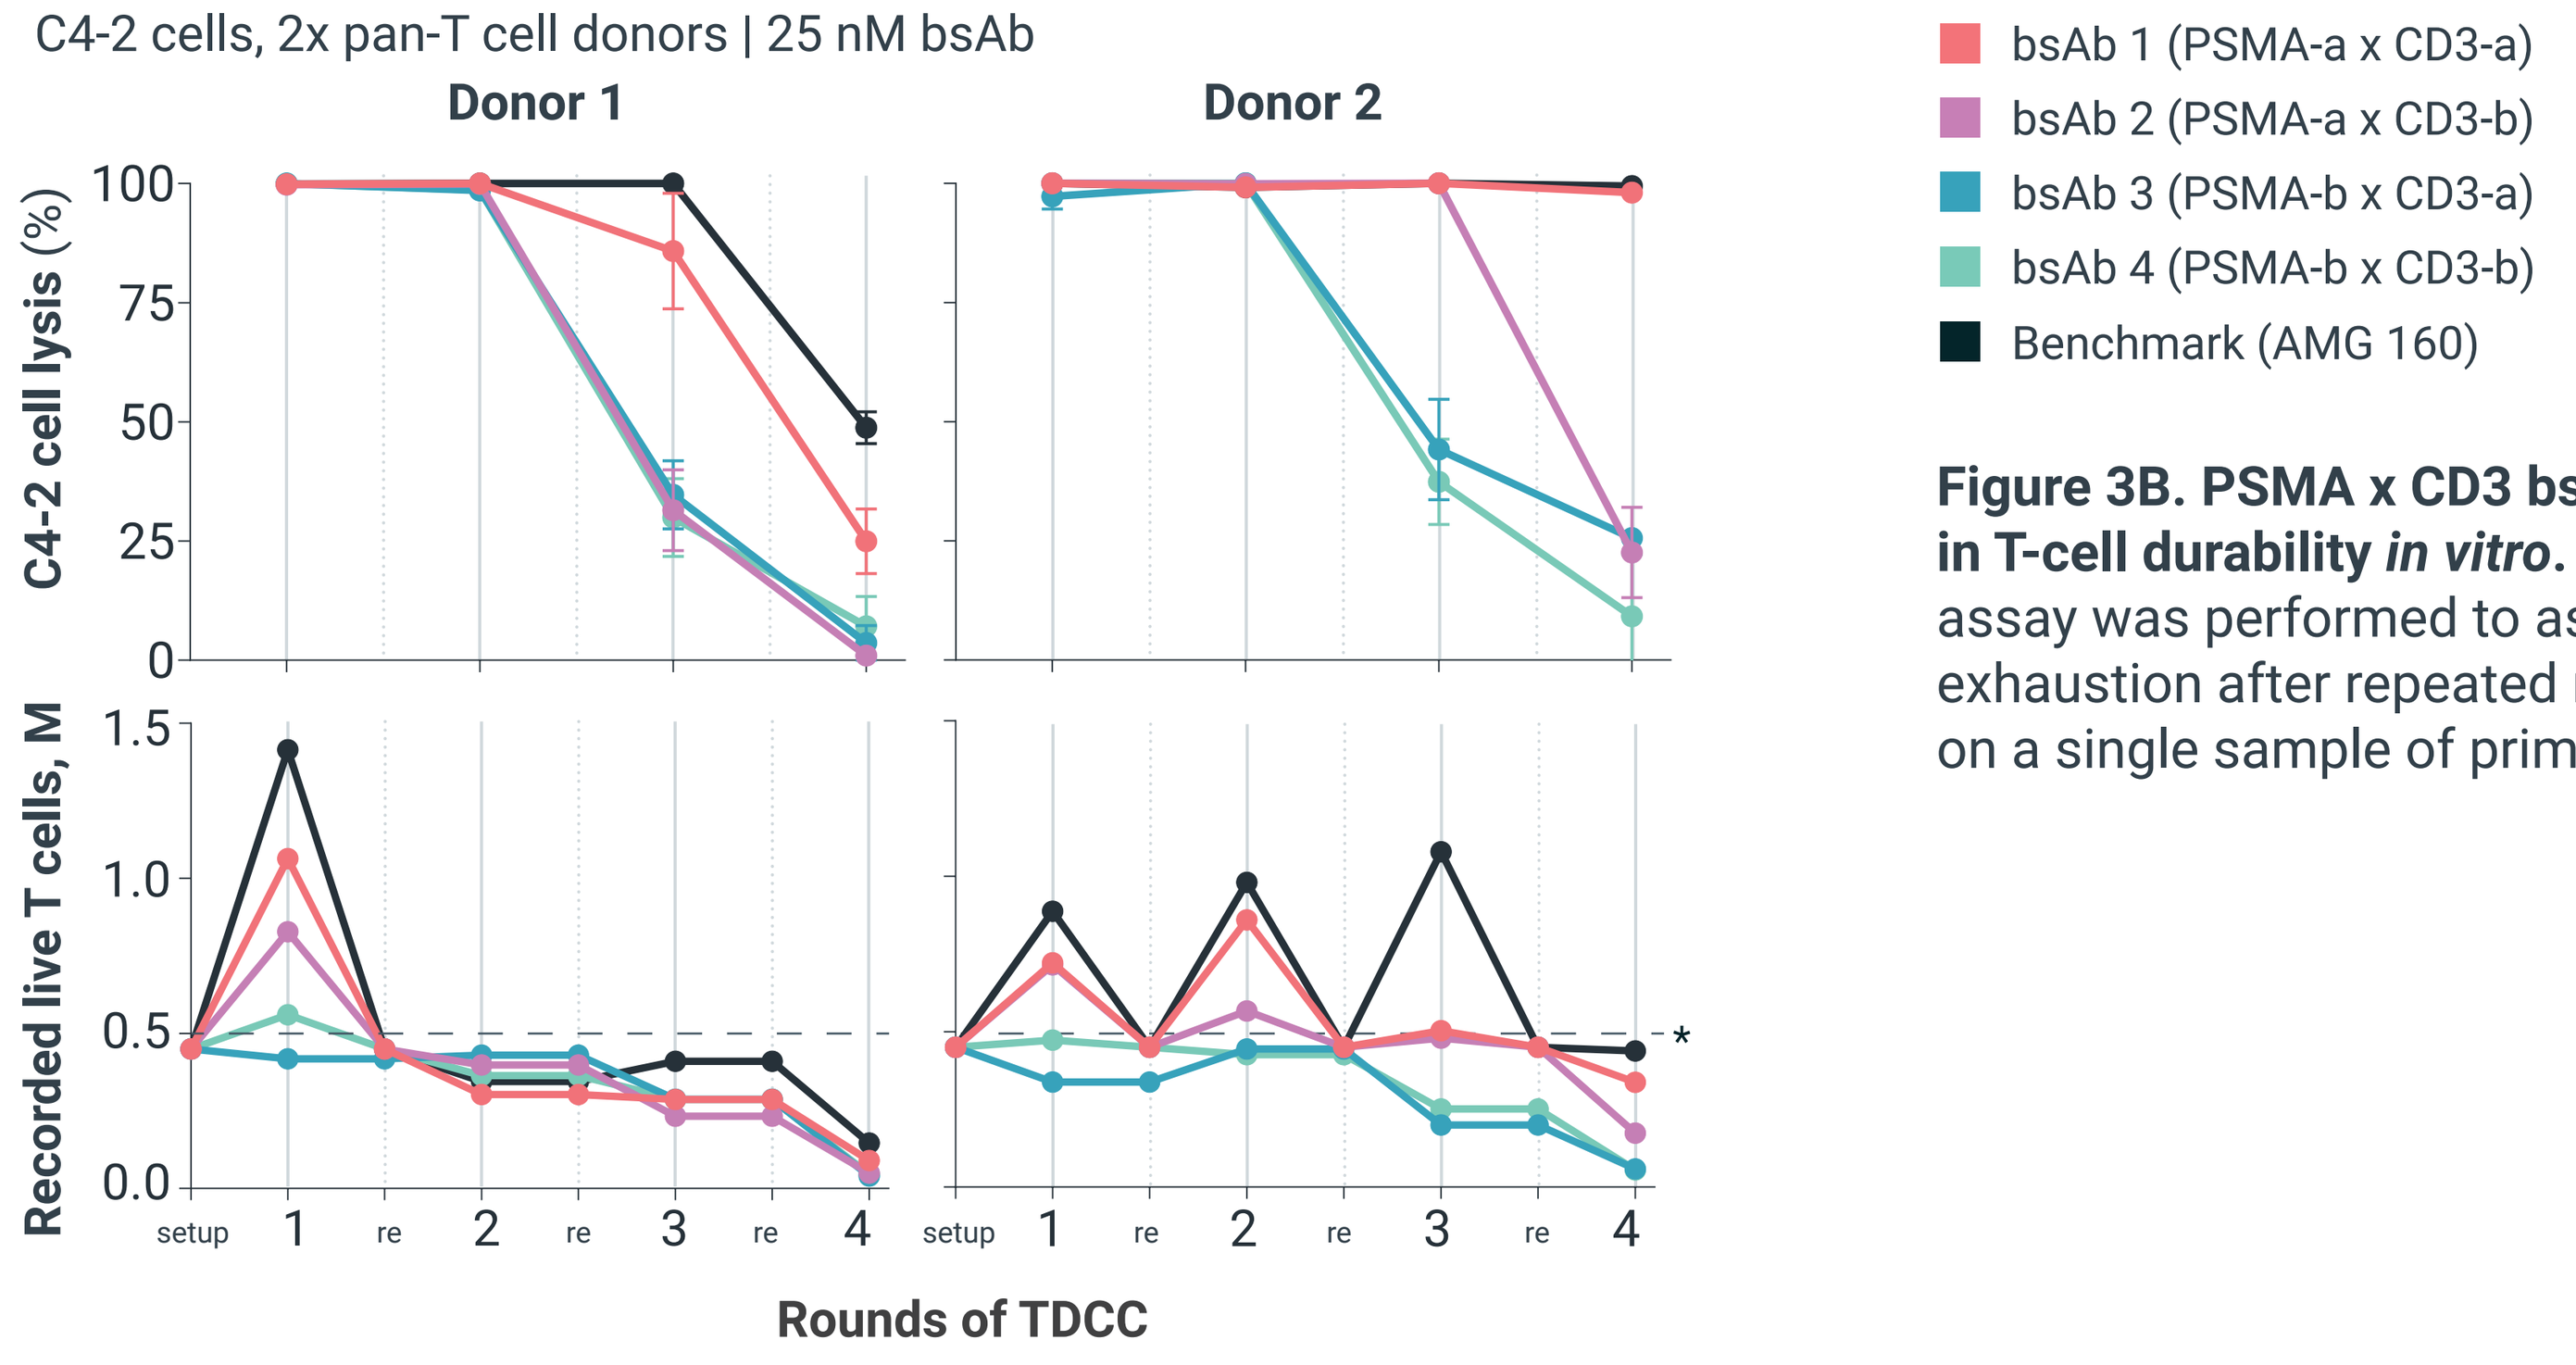
C4-2 cells, 2x pan-T cell donors | 25 nM bsAb
Donor 1
Donor 2
C4-2 cell lysis (%)
Recorded live T cells, M
Rounds of TDCC
Bispecifics
bsAb 1 (PSMA-a x CD3-a)
bsAb 2 (PSMA-a x CD3-b)
bsAb 3 (PSMA-b x CD3-a)
bsAb 4 (PSMA-b x CD3-b)
Benchmark (AMG 160)

Figure 3B. PSMA x CD3 bsAbs show variation in T-cell durability *in vitro*. A serial T-cell killing assay was performed to assess T-cell exhaustion after repeated rounds of TDCC on a single sample of primary T cells.

PSMA x CD3 bispecifics show robust anti-tumor efficacy *in vivo*

TCEs show tumor growth inhibition across multiple dose levels and tumor models

Tumor volume

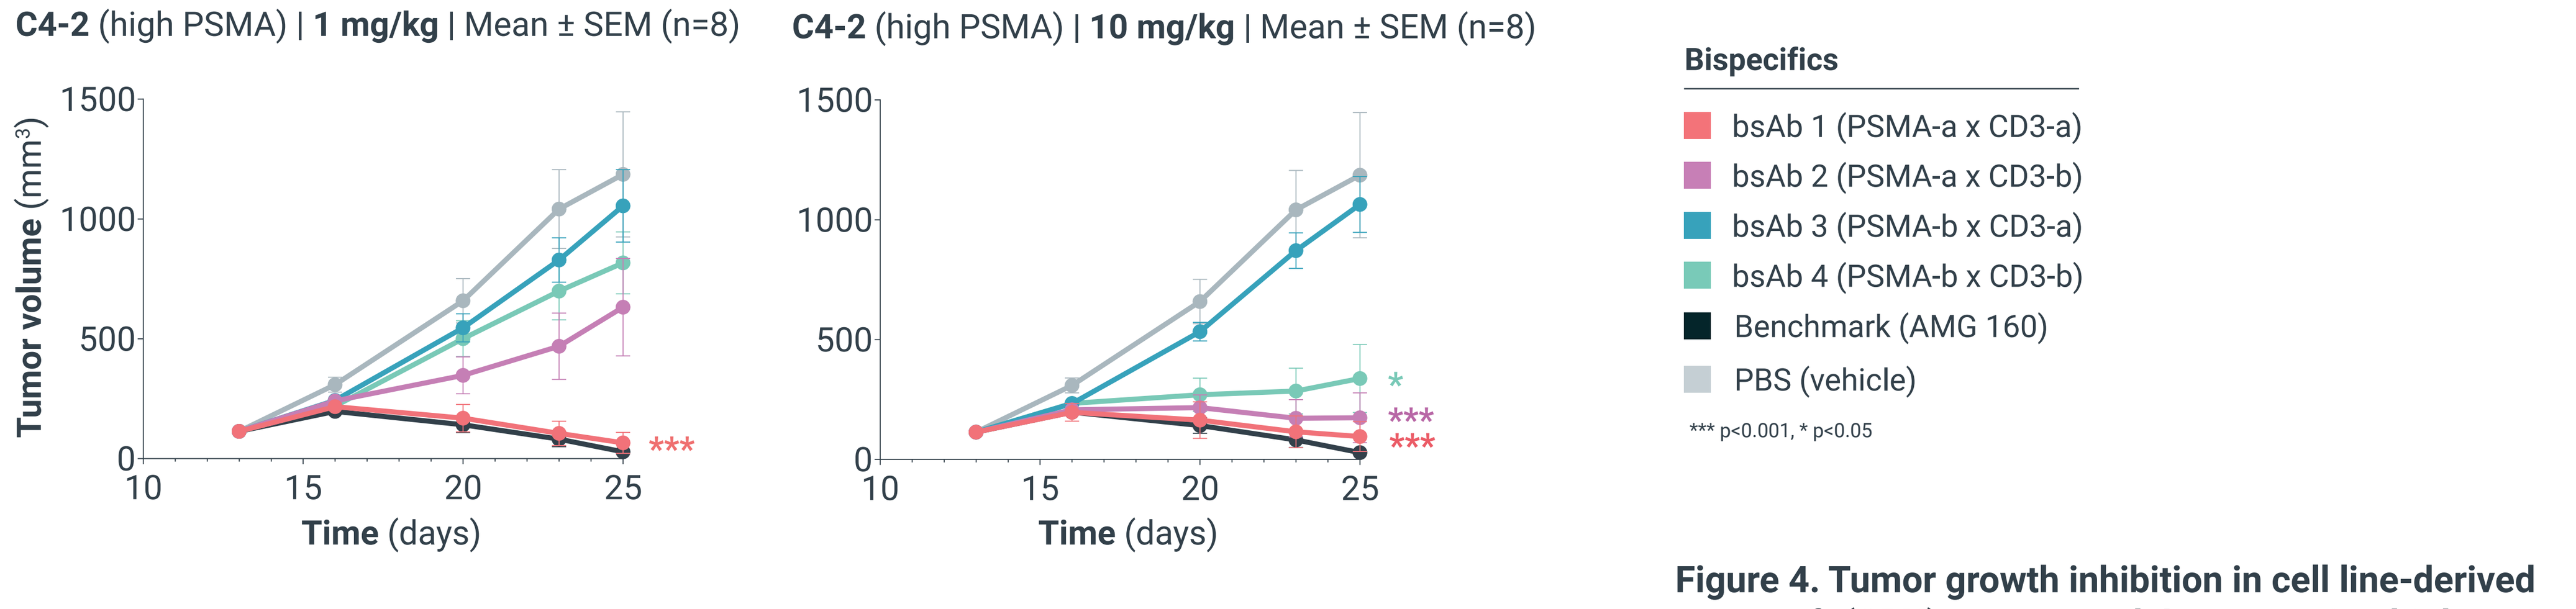
C4-2 (high PSMA) | 1 mg/kg | Mean ± SEM (n=8)
C4-2 (high PSMA) | 10 mg/kg | Mean ± SEM (n=8)
Tumor volume (mm³)
Time (days)

*
Bispecifics
bsAb 1 (PSMA-a x CD3-a)
bsAb 2 (PSMA-a x CD3-b)
bsAb 3 (PSMA-b x CD3-a)
bsAb 4 (PSMA-b x CD3-b)
Benchmark (AMG 160)
PBS (vehicle)
*** p<0.001, * p<0.05

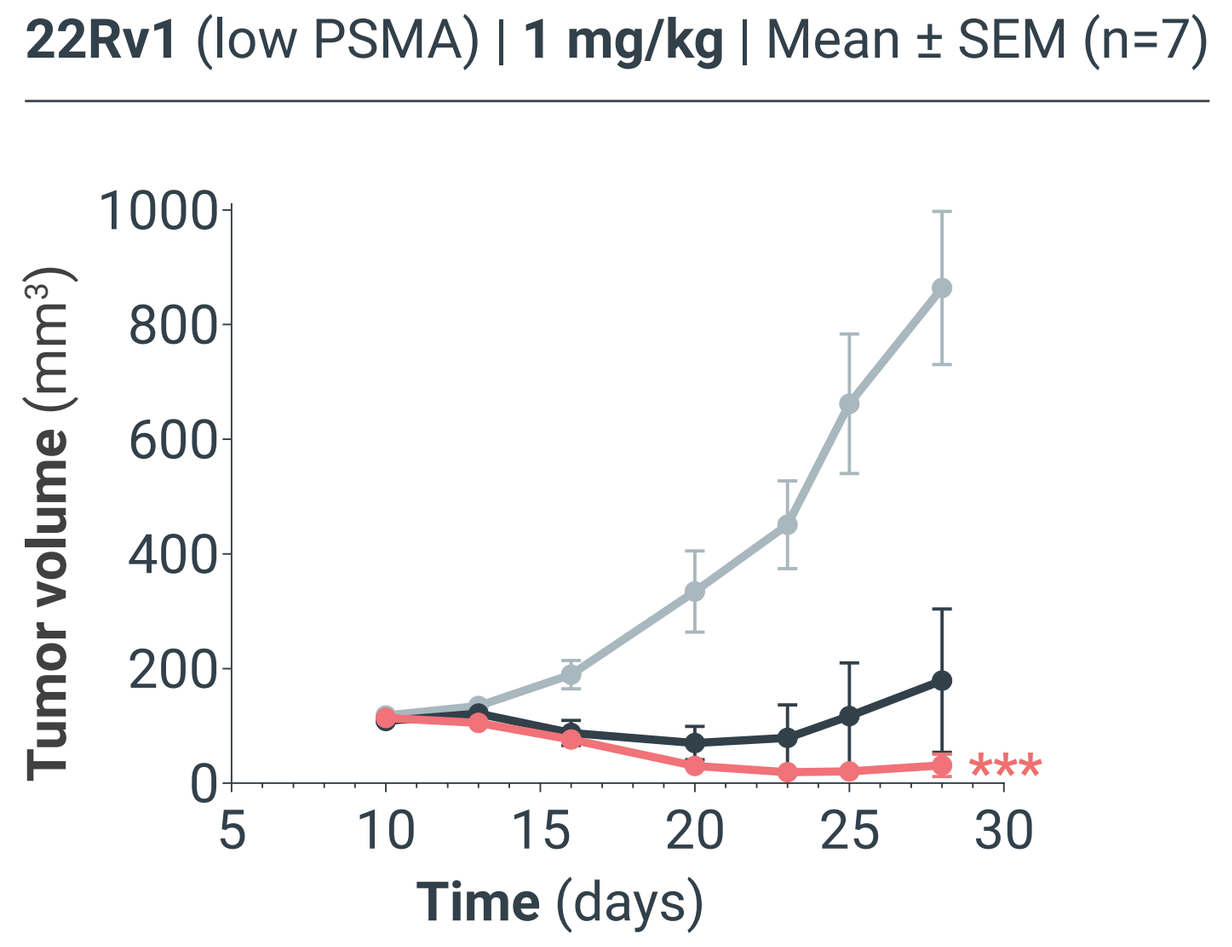
22Rv1 (low PSMA) | 1 mg/kg | Mean ± SEM (n=7)
Tumor volume (mm³)
Time (days)

Tumor xenograft mouse model

In vivo experiment schematic

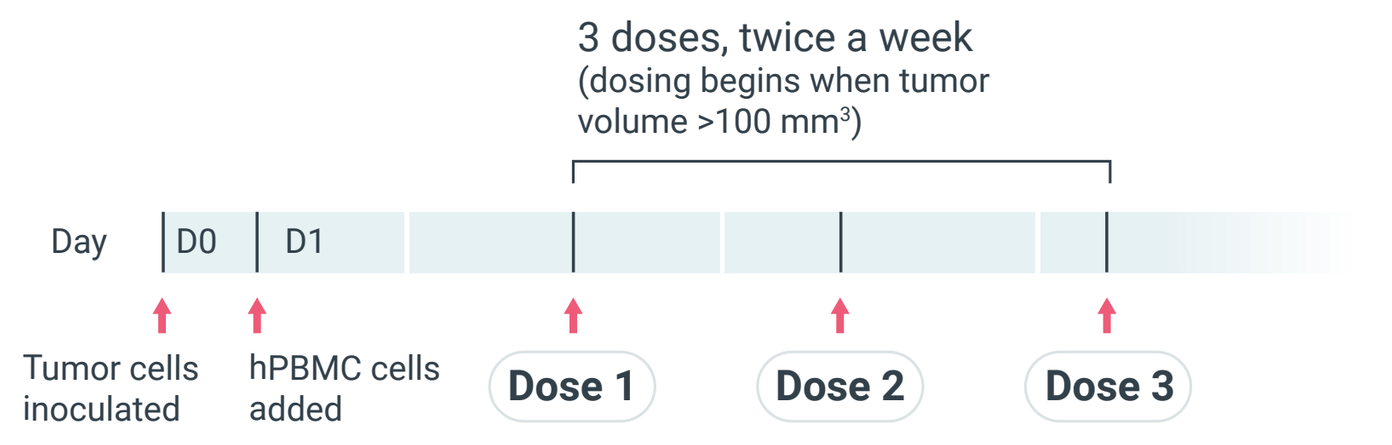
3 doses, twice a week (dosing begins when tumor volume >100 mm³)
Day
D0
D1
Tumor cells inoculated
hPBMC cells added
Dose 1
Dose 2
Dose 3

Figure 4. Tumor growth inhibition in cell line-derived xenograft (CDX) mouse models expressing high (C4-2) and low (22Rv1) levels of PSMA. Molecules harboring novel CD3 arms show dose-responsive anti-tumor efficacy, with enhanced tumor regression for bsAb 2 and 4 at higher dosage. Statistical significance of tumor volume reduction compared to PBS control at day 25 was assessed using ANOVA. The benchmark molecule, AMG 160, was dosed at 0.07 and 0.7 mg/kg in the C4-2 and 22Rv1 tumor models, respectively.

Conclusions

Multiple TCE parameters impact function

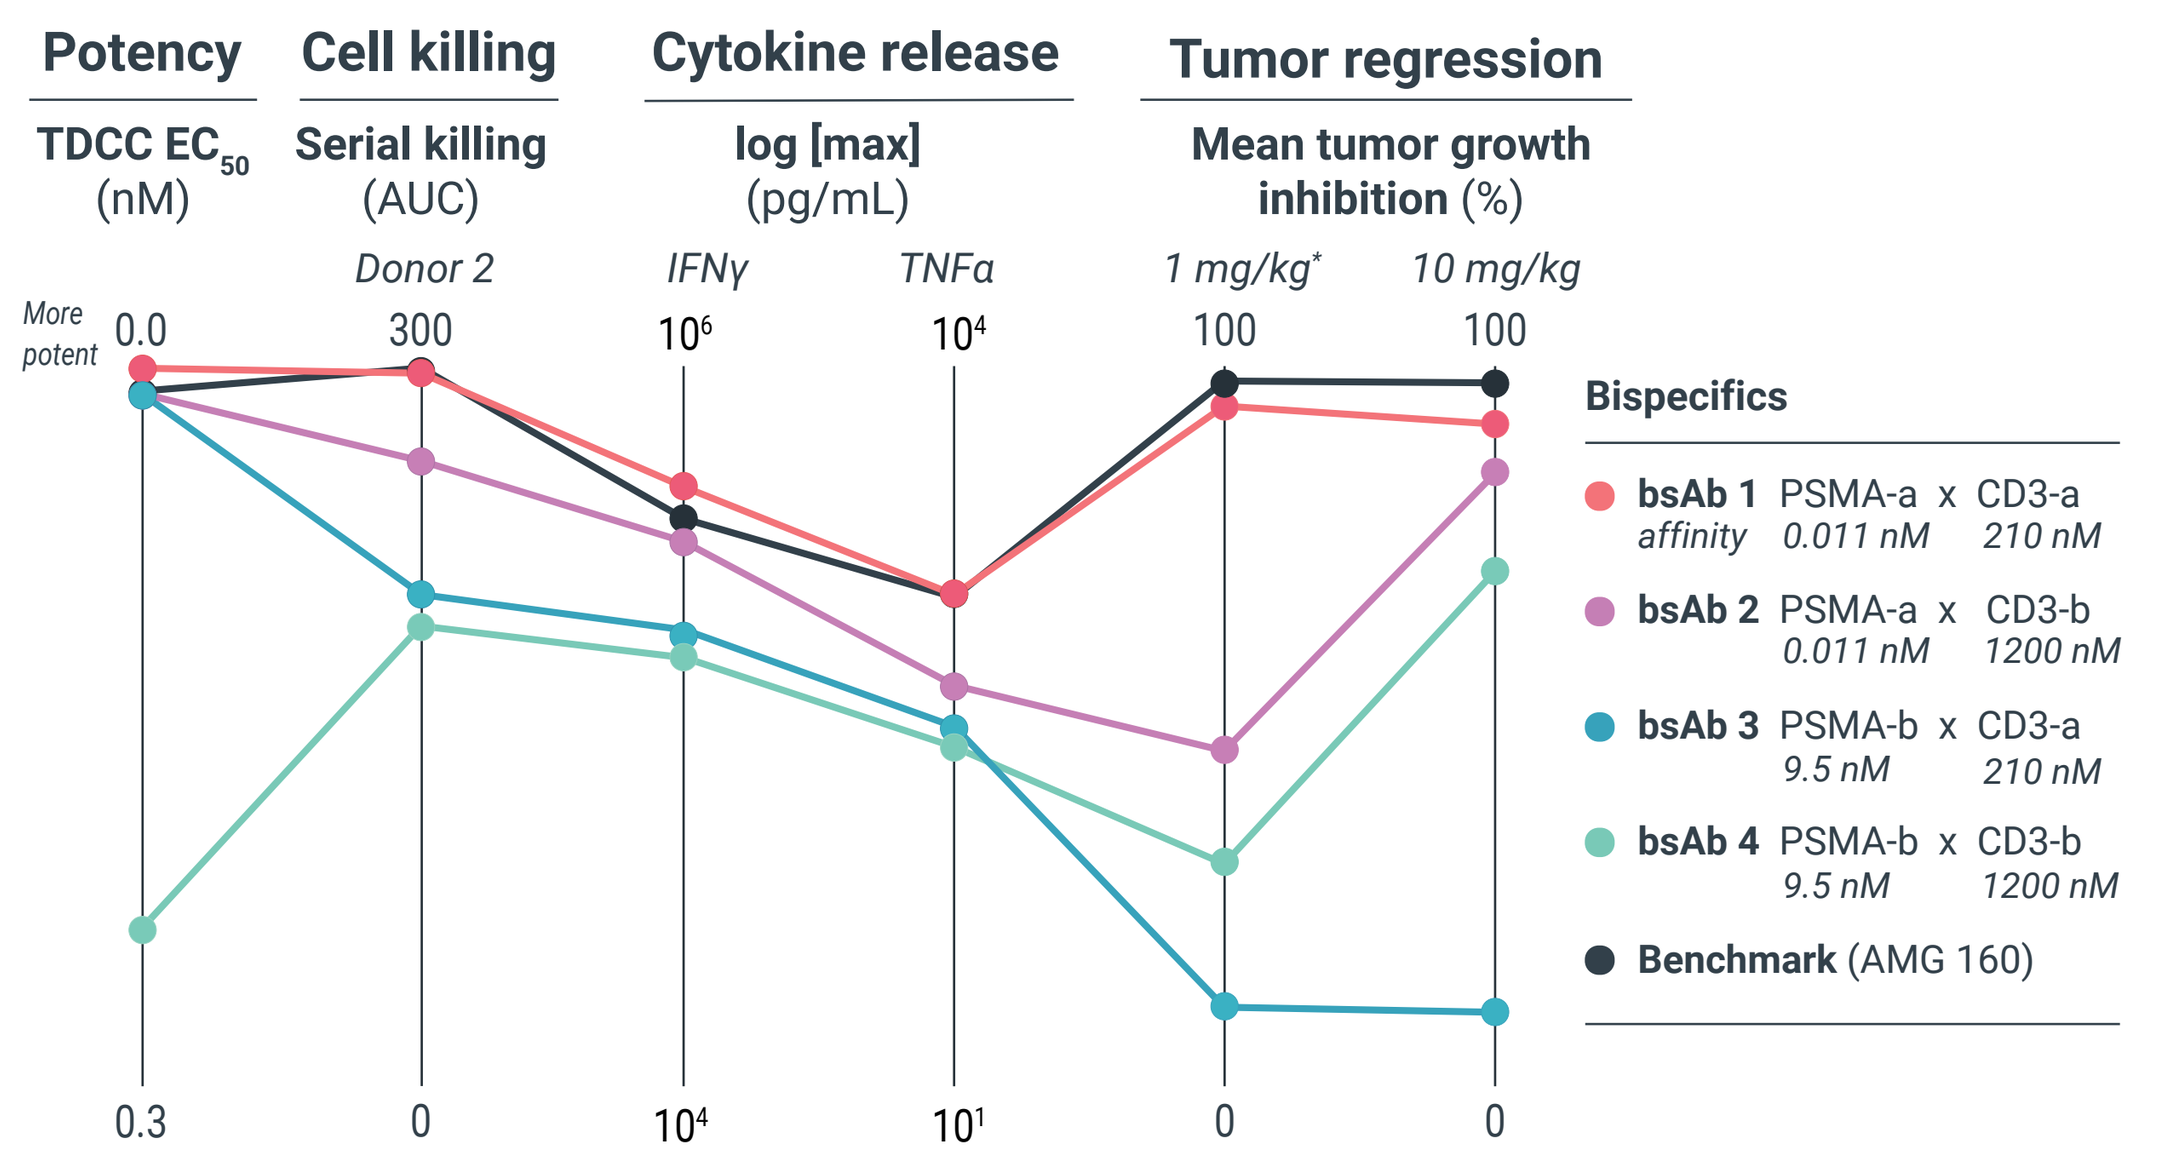
Potency
TDCC EC50 (nM)
Cell killing
Serial killing (AUC)
Donor 2
Cytokine release
log [max] (pg/mL)
IFNγ
TNFα
Tumor regression
Mean tumor growth inhibition (%)
1 mg/kg*
10 mg/kg
More potent
Bispecifics
bsAb 1 PSMA-a x CD3-a
affinity 0.011 nM 210 nM
bsAb 2 PSMA-a x CD3-b
0.011 nM 1200 nM
bsAb 3 PSMA-b x CD3-a
9.5 nM 210 nM
bsAb 4 PSMA-b x CD3-b
9.5 nM 1200 nM
Benchmark (AMG 160)

Figure 5. PSMA x CD3 bsAbs with distinct epitopes and affinities show unique functional profiles *in vitro* and *in vivo*. Summary of functional characterization data, TDCC axis has been inverted to support data interpretation and serial killing area under the curve (AUC) was calculated from a plot of C4-2 lysis (%) against rounds of TDCC for a representative donor.

*AMG 160 was dosed at 0.07 mg/kg in tumor growth inhibition studies (C4-2 model).

Costimulatory strategies to enhance potency

Serial T-cell killing

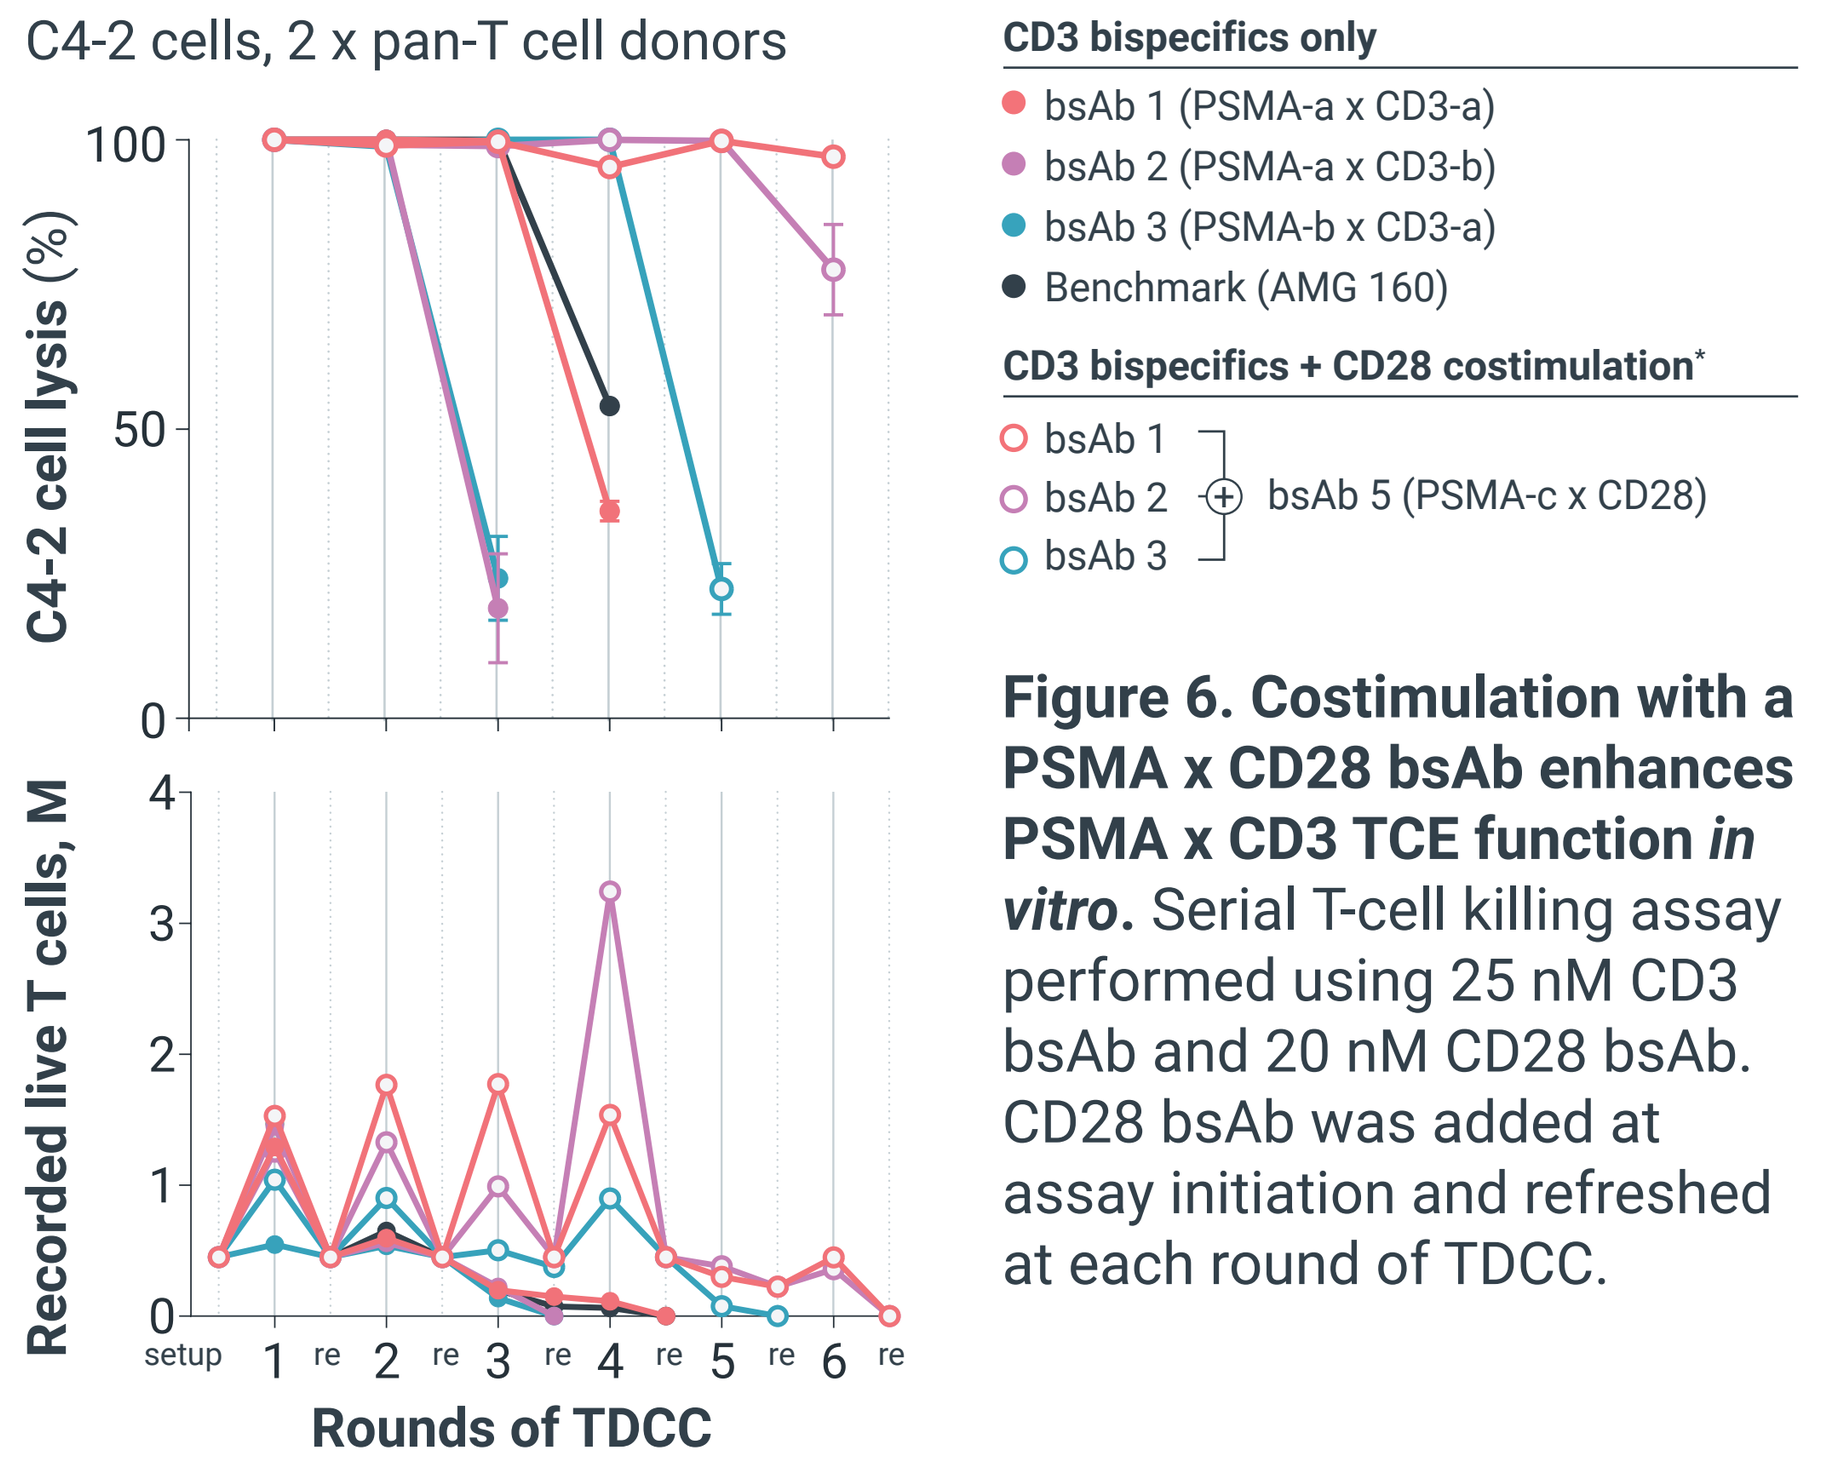
C4-2 cells, 2 x pan-T cell donors
C4-2 cell lysis (%)
Recorded live T cells, M
Rounds of TDCC
CD3 bispecifics only
bsAb 1 (PSMA-a x CD3-a)
bsAb 2 (PSMA-a x CD3-b)
bsAb 3 (PSMA-b x CD3-a)
Benchmark (AMG 160)
CD3 bispecifics + CD28 costimulation*
bsAb 1
bsAb 2
bsAb 3
bsAb 5 (PSMA-c x CD28)

Figure 6. Costimulation with a PSMA x CD28 bsAb enhances PSMA x CD3 TCE function *in vitro*. Serial T-cell killing assay performed using 25 nM CD3 bsAb and 20 nM CD28 bsAb. CD28 bsAb was added at assay initiation and refreshed at each round of TDCC.

*The PSMA x CD28 bispecific was engineered using a published CD28 antibody sequence and an AbCellera-generated PSMA-binder (PSMA-c) that interacts with an epitope distinct from that of PSMA-a/b.

SUMMARY

Molecular diversity and in-depth preclinical assessment are needed to optimize TCE efficacy

- Diverse pairings of PSMA- and CD3-binders generate unique differences in functional activity
- In-depth, empirical testing is required to reveal how TCE parameters impact function and to optimize the immune synapse

NEXT STEPS

CD28-targeted costimulation enhances TCE anti-tumor activity

- Proof-of-concept data shows that PSMA x CD28 bsAbs enhance efficacy of TCEs by improving T-cell durability
- Assessment of novel bi- and multispecific costimulatory TCEs is ongoing